Single-cell RNA sequencing of paclitaxel-treated breast cancer cell lines to find individual cell response

Charles J Vaske[1], Wendy Lee[2], Stephen C Benz[1], John Z Sanborn[1], Fernando Lopez-Diaz[3], Beverly M Emerson[3], and Nader Pourmand[2]

[1]Five3 Genomics, LLC, Santa Cruz, CA, United States, 95060;
[2]Biomolecular Engineering, University of California Santa Cruz, Santa Cruz, CA, United States, 95064 and
[3]Salk Institute, La Jolla, CA, United States, 92037

Abstract #1229

San Antonio Breast Cancer Symposium– December 4-8, 2012

Abstract

Cancer treatments act on a population of cells, each of which may experience different individual responses to treatment. Such differential response will result in resistance to treatment even if a majority of cancerous cells are eliminated. To examine differential cell response, we simultaneously profiled the gene expression and mutation spectrum of individual cells from the MDAMB231 cell line using next generation sequencing of isolated RNA. A total of 23 transcriptomes were characterized from paclitaxel-treated and paclitaxel-surviving cells. We found significant different changes in mutation rates between paclitaxel treated cells, with a dose-dependent increase in single nucleotide changes in RNA in paclitaxel-treated cells. Cells undergoing exposure to paclitaxel also showed higher pathway activity in SRC, as well as an integrin switch from ITGB1 to ITGB3. In contrast, cells that survived a high dose of paclitaxel showed an insignificant number of single nucleotide changes, suggesting that these cells either evaded initial paclitaxel exposure or were better able to repair the effects of paclitaxel exposure. Despite the RNA sequence similarity between surviving and untreated cells, there were changes in gene expression and pathway activities including higher PI3K activity. Paclitaxel-surviving cells also showed activation of pathways associated with higher proliferation.

Taxol Treatment Scheme

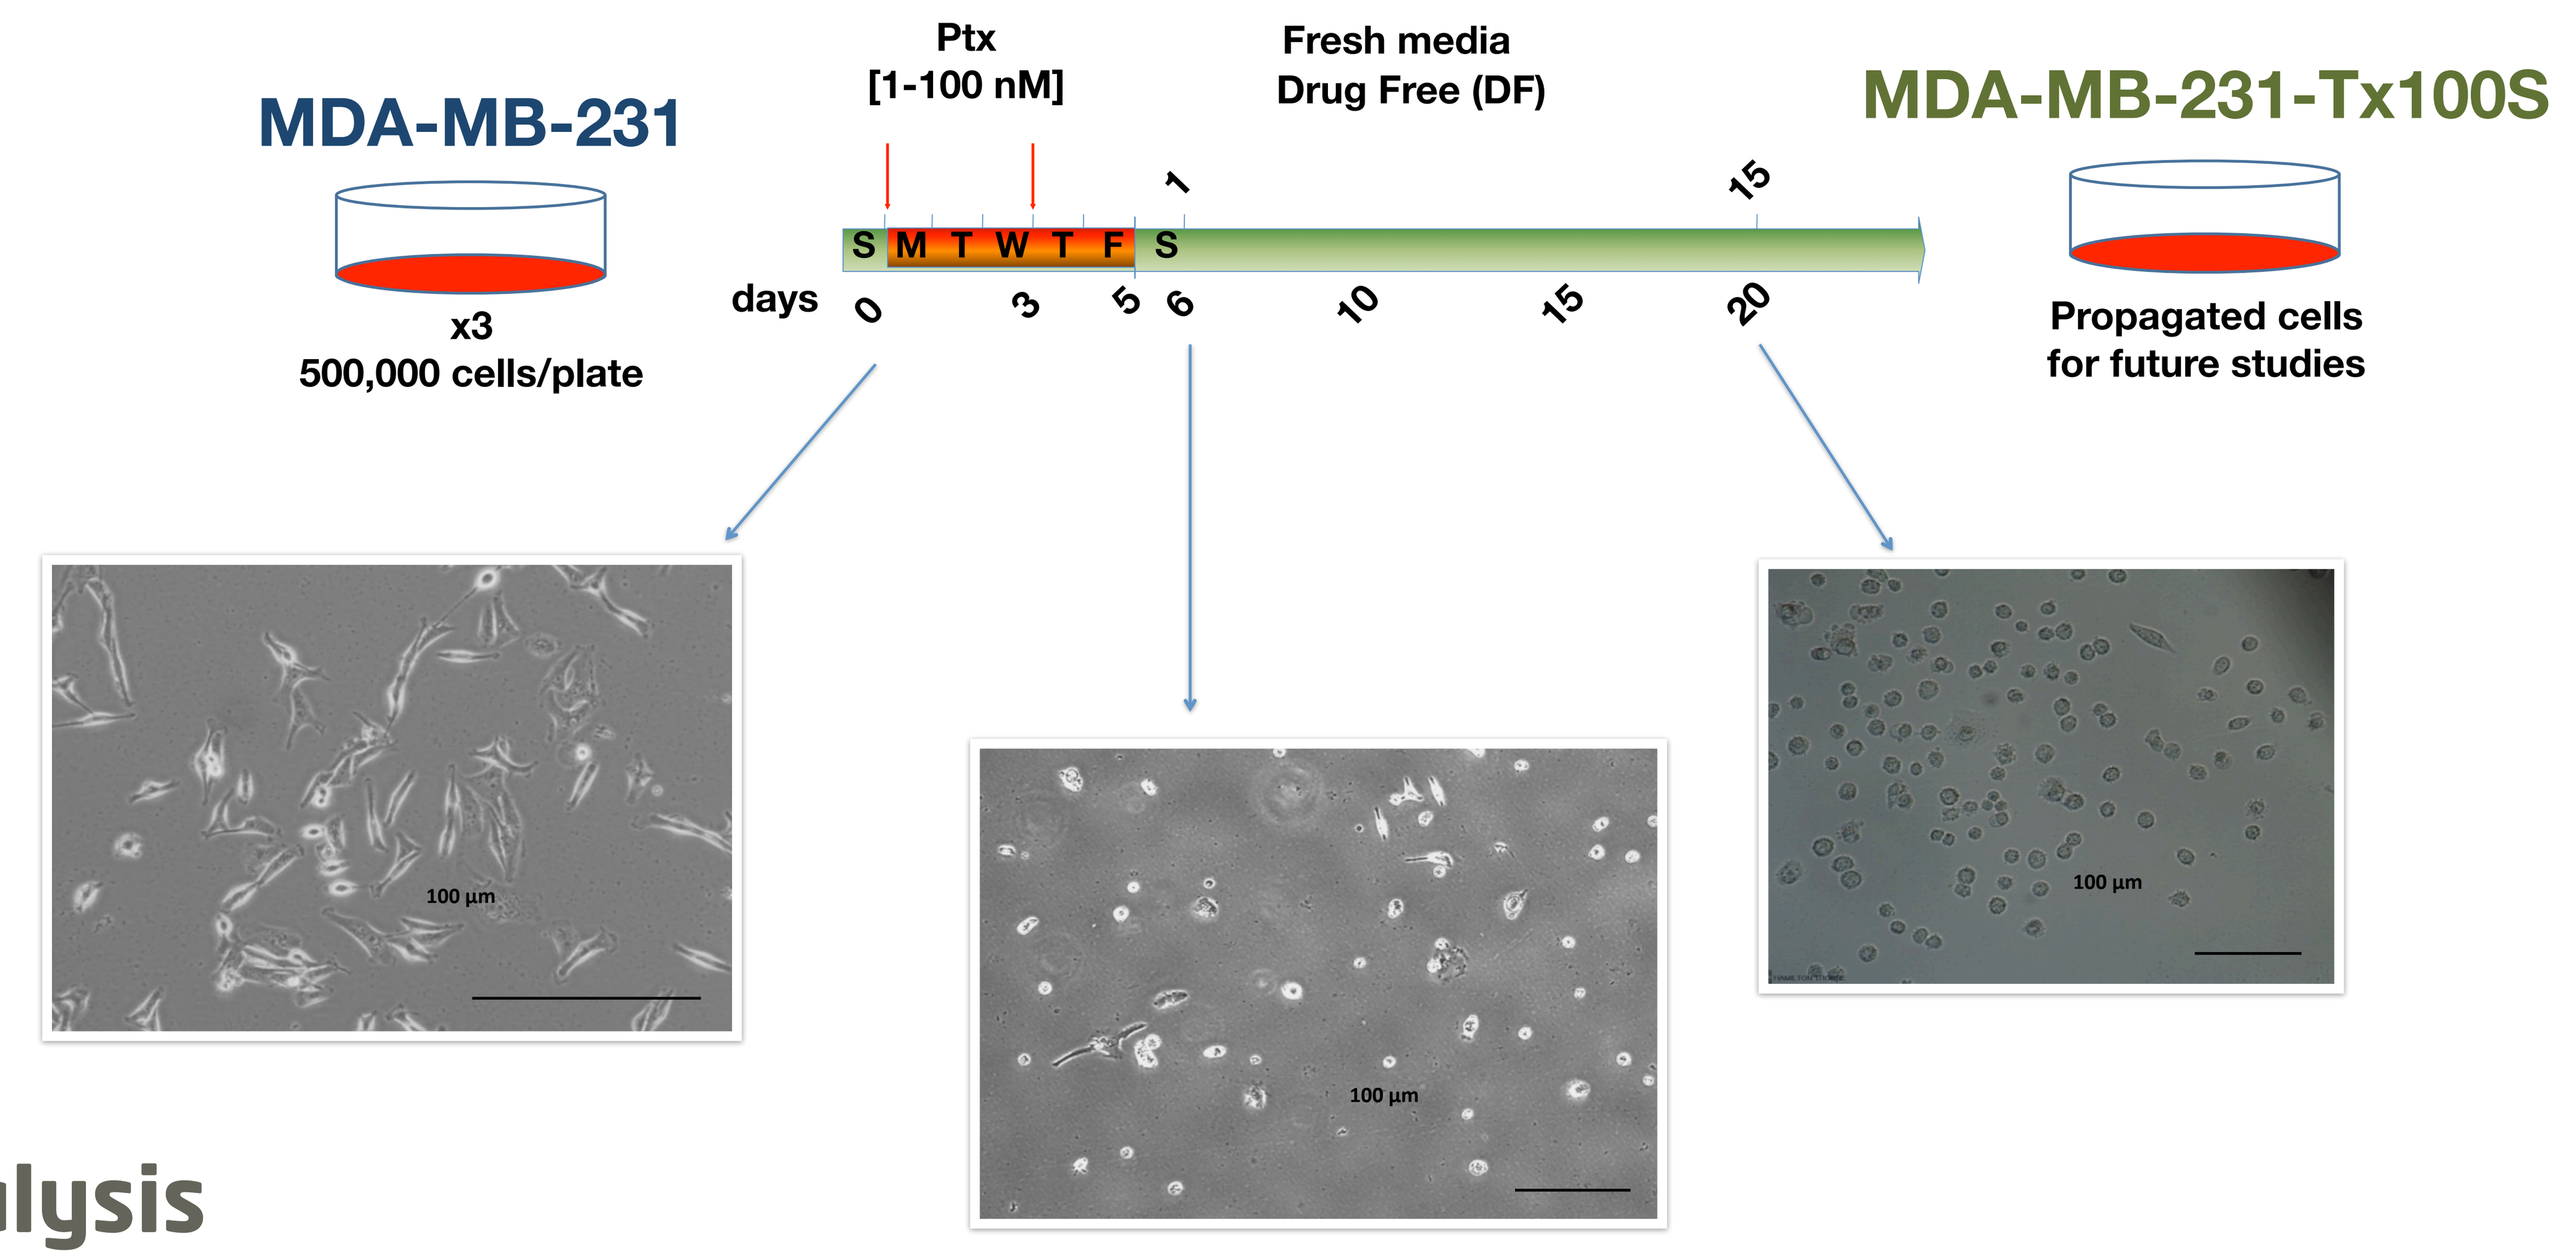
MDA-MB-231
x3
500,000 cells/plate
Ptx
[1-100 nM]
Fresh media
Drug Free (DF)
S M T W T F S
days
0
3
5
6
10
15
20
1
15
MDA-MB-231-Tx100S
Propagated cells
for future studies
100 µm
100 µm
100 µm

Single-cell RNA Expression and Pathway Analysis

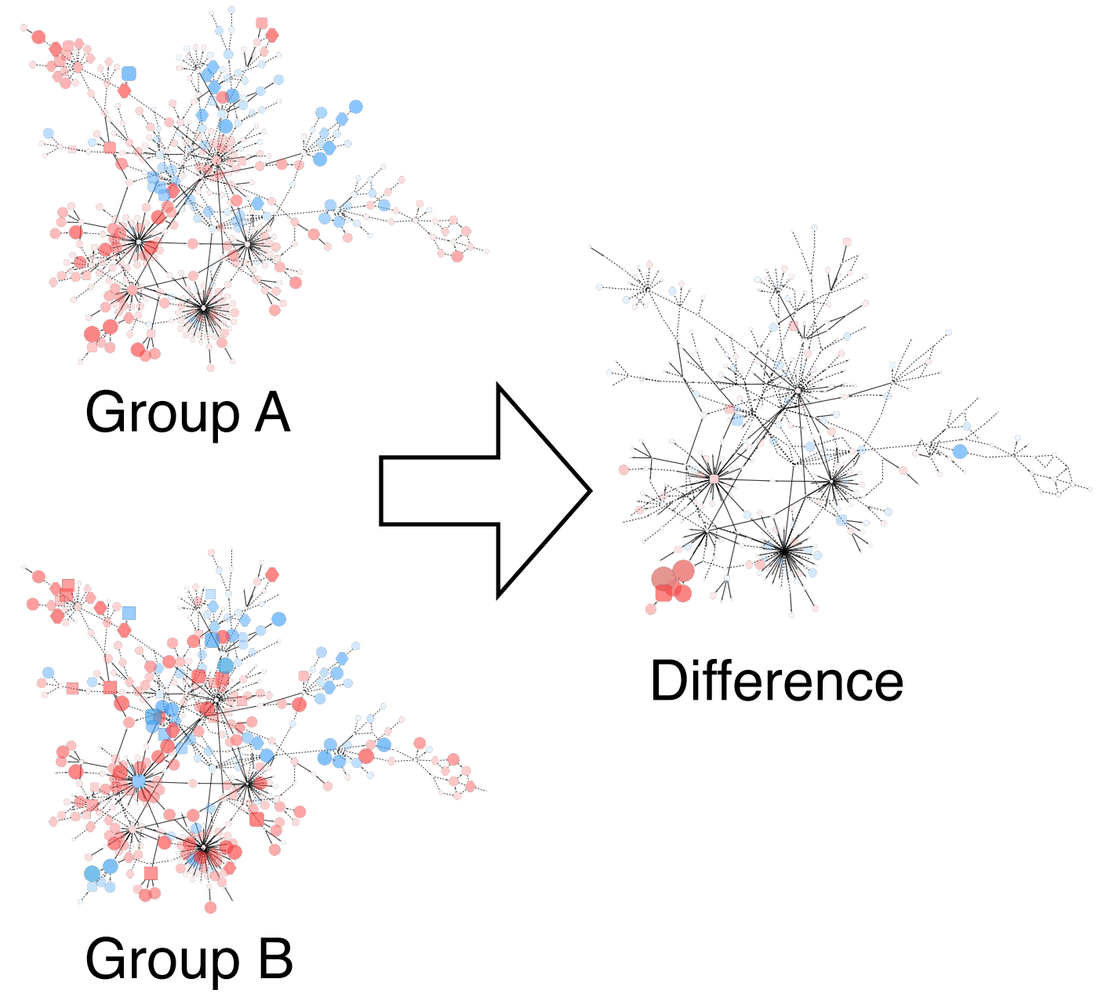
Group A
Group B
Difference

Using the Paradigm pathway analysis method, differential pathway activities were extracted from the three treatment groups. Paradigm was able to find statistically significant differential sub-pathways in each condition comparison, something that challenged traditional RNA-seq differential expression methods such as DEseq.

Treated cells show a switch from Integrin Beta 1 to Integrin Beta 3, however Beta 3 is lost in the survivors. Survivors show increased G2/M transition as well as generally higher proliferation, concordant with a resumption of cell cycle after stalling.

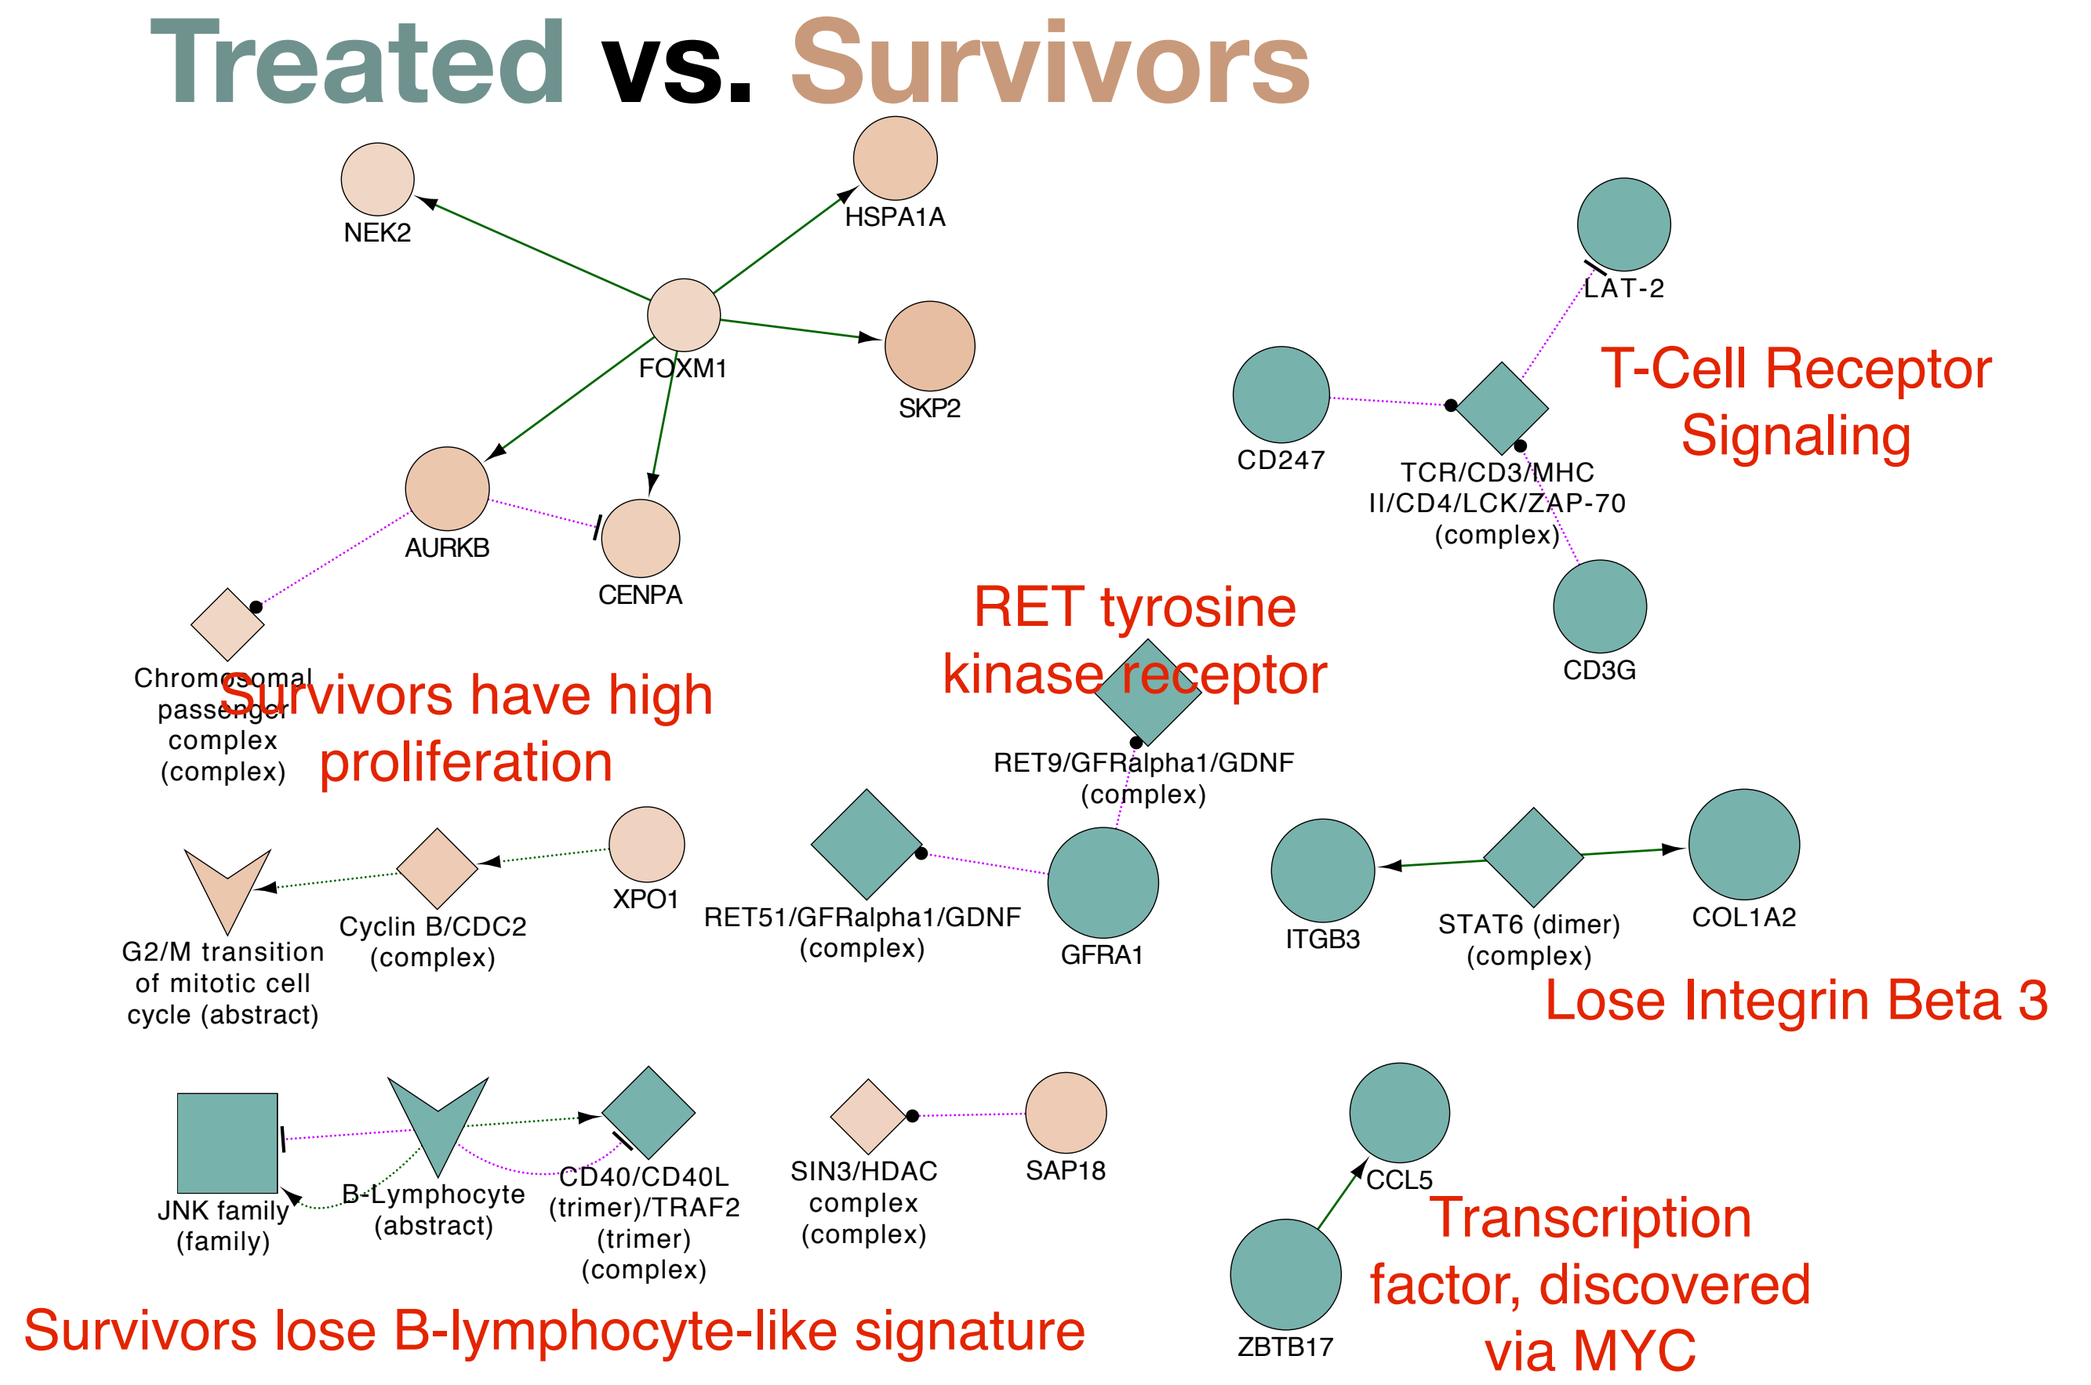
Treated vs. Survivors
NEK2
HSPA1A
FOXM1
SKP2
AURKB
CENPA
Survivors have high proliferation
RET tyrosine kinase receptor
T-Cell Receptor Signaling
CD247
LAT-2
CD3G
RET9/GFRalpha1/GDNF (complex)
RET51/GFRalpha1/GDNF (complex)
GFRA1
ITGB3
STAT6 (dimer) (complex)
COL1A2
Lose Integrin Beta 3
G2/M transition of mitotic cell cycle (abstract)
Cyclin B/CDC2 (complex)
XPO1
JNK family (family)
B-Lymphocyte (abstract)
CD40/CD40L (trimer)/TRAF2 (trimer) (complex)
SIN3/HDAC complex (complex)
SAP18
CCL5
ZBTB17
Transcription factor, discovered via MYC
Survivors lose B-lymphocyte-like signature

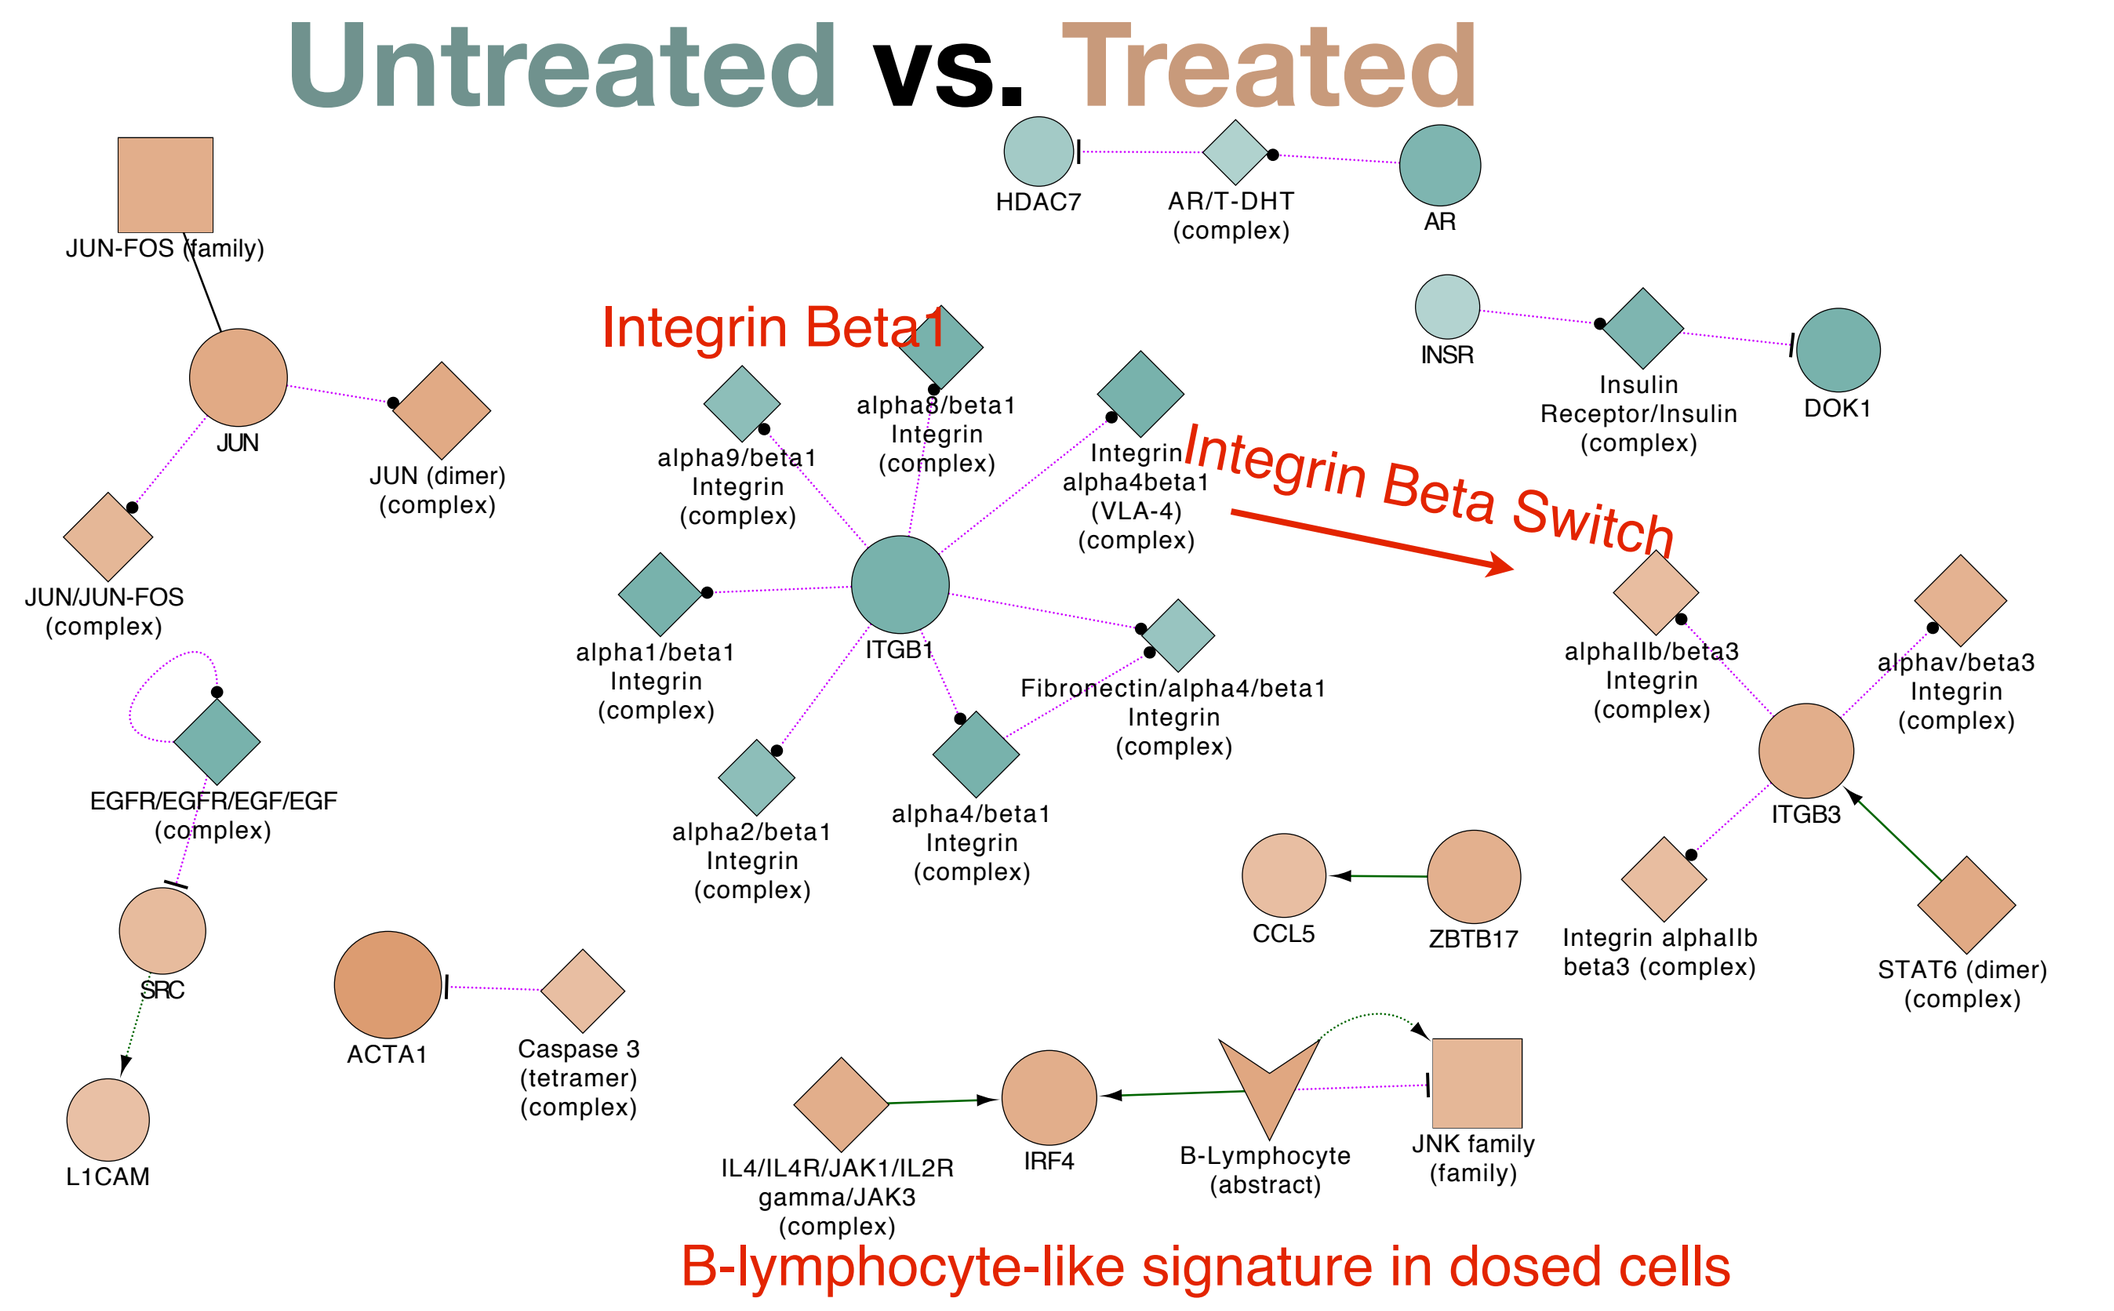
Untreated vs. Treated
JUN-FOS (family)
JUN
JUN (dimer) (complex)
JUN/JUN-FOS (complex)
EGFR/EGFR/EGF/EGF (complex)
SRC
L1CAM
ACTA1
Caspase 3 (tetramer) (complex)
Integrin Beta1
ITGB1
Integrin Beta Switch
ITGB3
HDAC7
AR/T-DHT (complex)
AR
INSR
Insulin Receptor/Insulin (complex)
DOK1
CCL5
ZBTB17
IRF4
B-Lymphocyte (abstract)
JNK family (family)
STAT6 (dimer) (complex)
B-lymphocyte-like signature in dosed cells

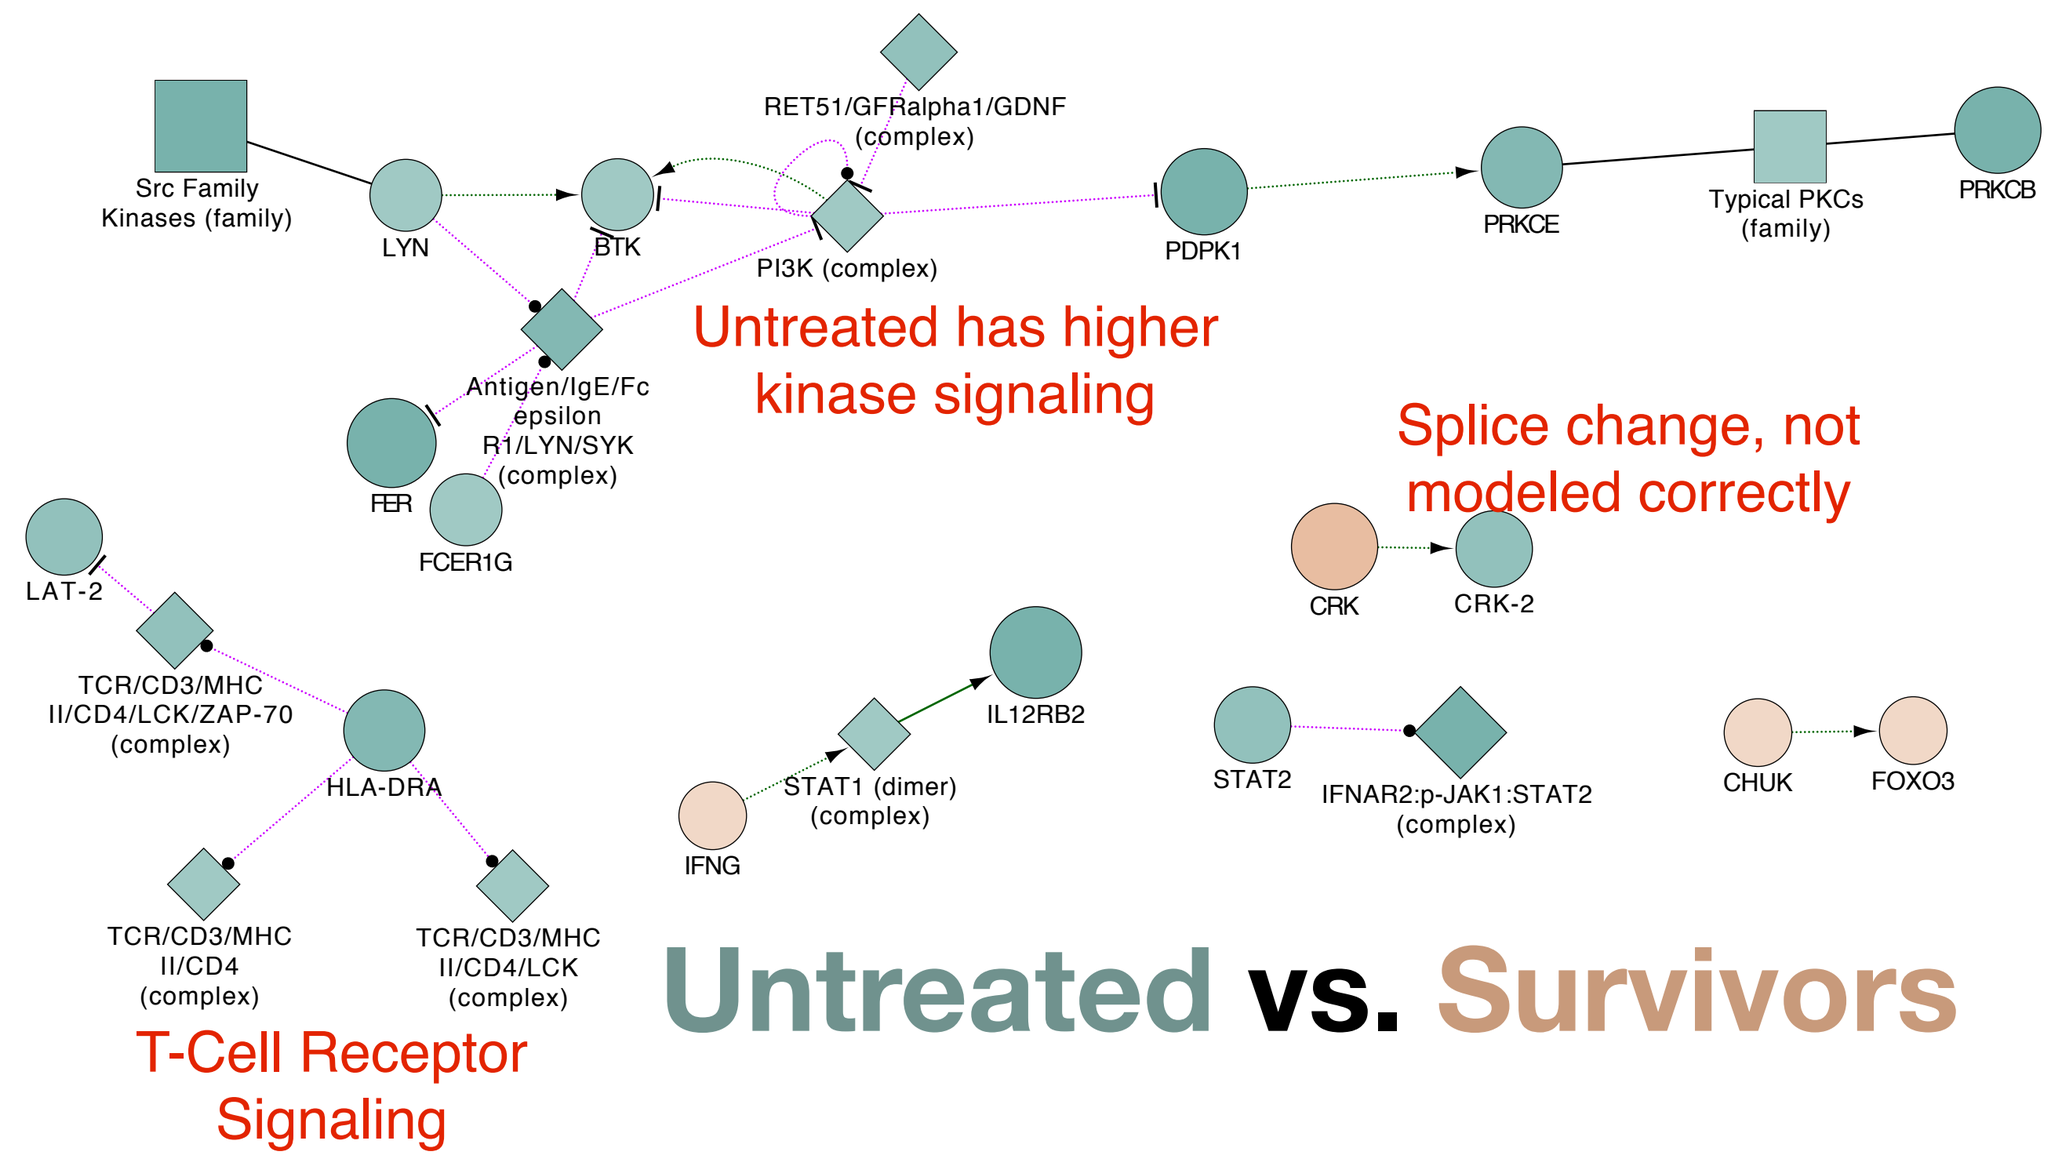
Src Family Kinases (family)
LYN
BTK
RET51/GFRalpha1/GDNF (complex)
PI3K (complex)
PDPK1
PRKCE
Typical PKCs (family)
PRKCB
Untreated has higher kinase signaling
Splice change, not modeled correctly
FER
FCER1G
LAT-2
HLA-DRA
IL12RB2
STAT1 (dimer) (complex)
IFNG
CRK
CRK-2
STAT2
IFNAR2:p-JAK1:STAT2 (complex)
CHUK
FOXO3
T-Cell Receptor Signaling
Untreated vs. Survivors

Single-cell RNA Sequencing Coverage

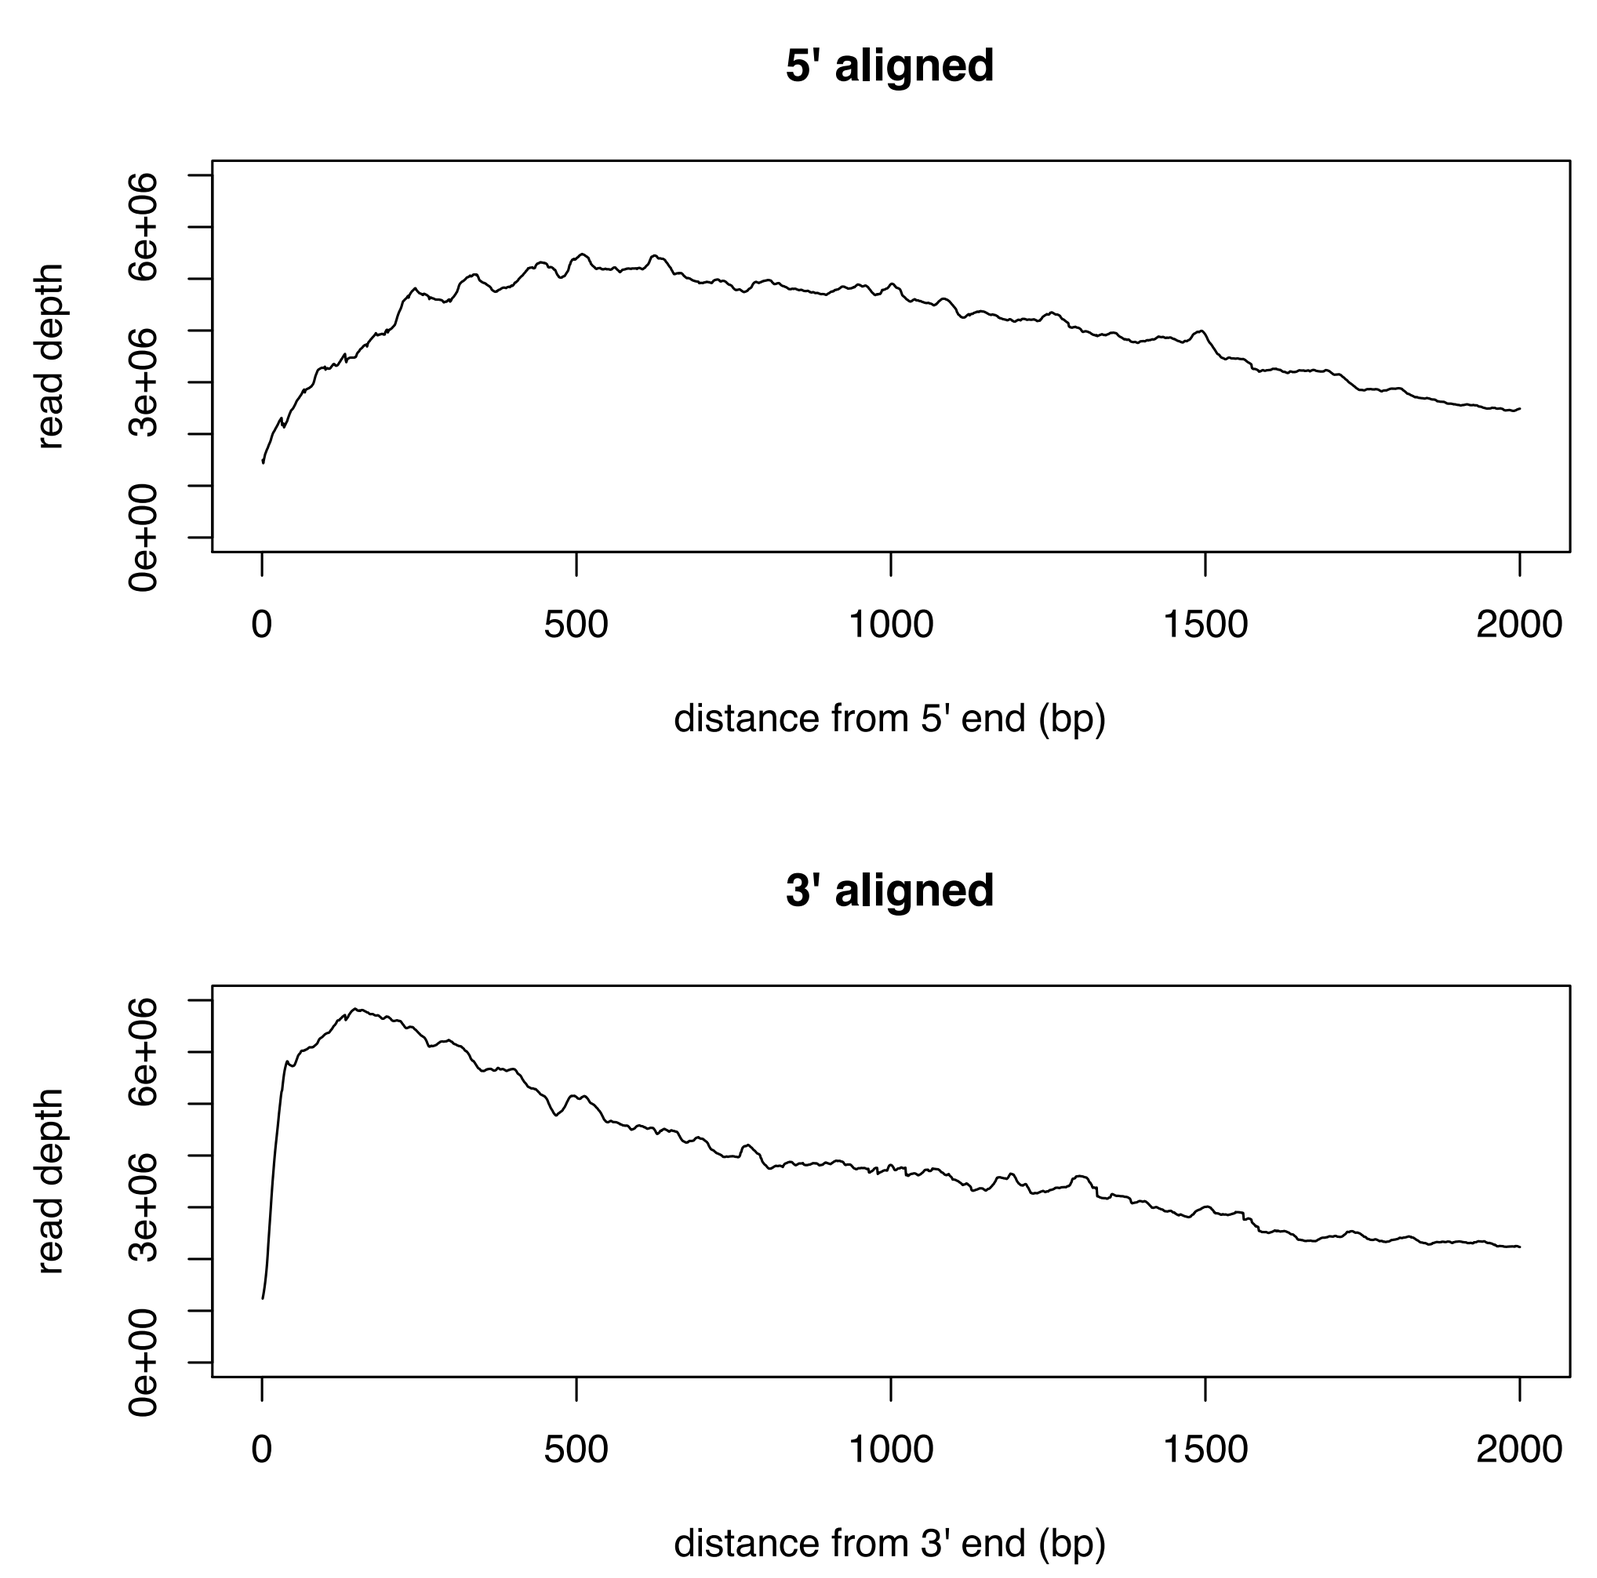
5' aligned
read depth
distance from 5' end (bp)
3' aligned
distance from 3' end (bp)
0e+00
3e+06
6e+06
0
500
1000
1500
2000

Aggregate RNA sequence coverage across known genes shows coverage across entire gene length. The top plot shows the total number of reads that cover any gene starting at the 5' end of genes. The bottom blot shows aggregate coverage starting from the 3' end of genes. There is a small 3' bias towards reads, but solid coverage even within the 5' end of genes.

Individual cell RNA sequence coverage shows which sections of mRNAs are amplified in the single cell preparation. Typical islands are 500+ bp in length. The coding length of CDK4 is 303 amino acids, while ATM has a coding length of 3056 amino acids.

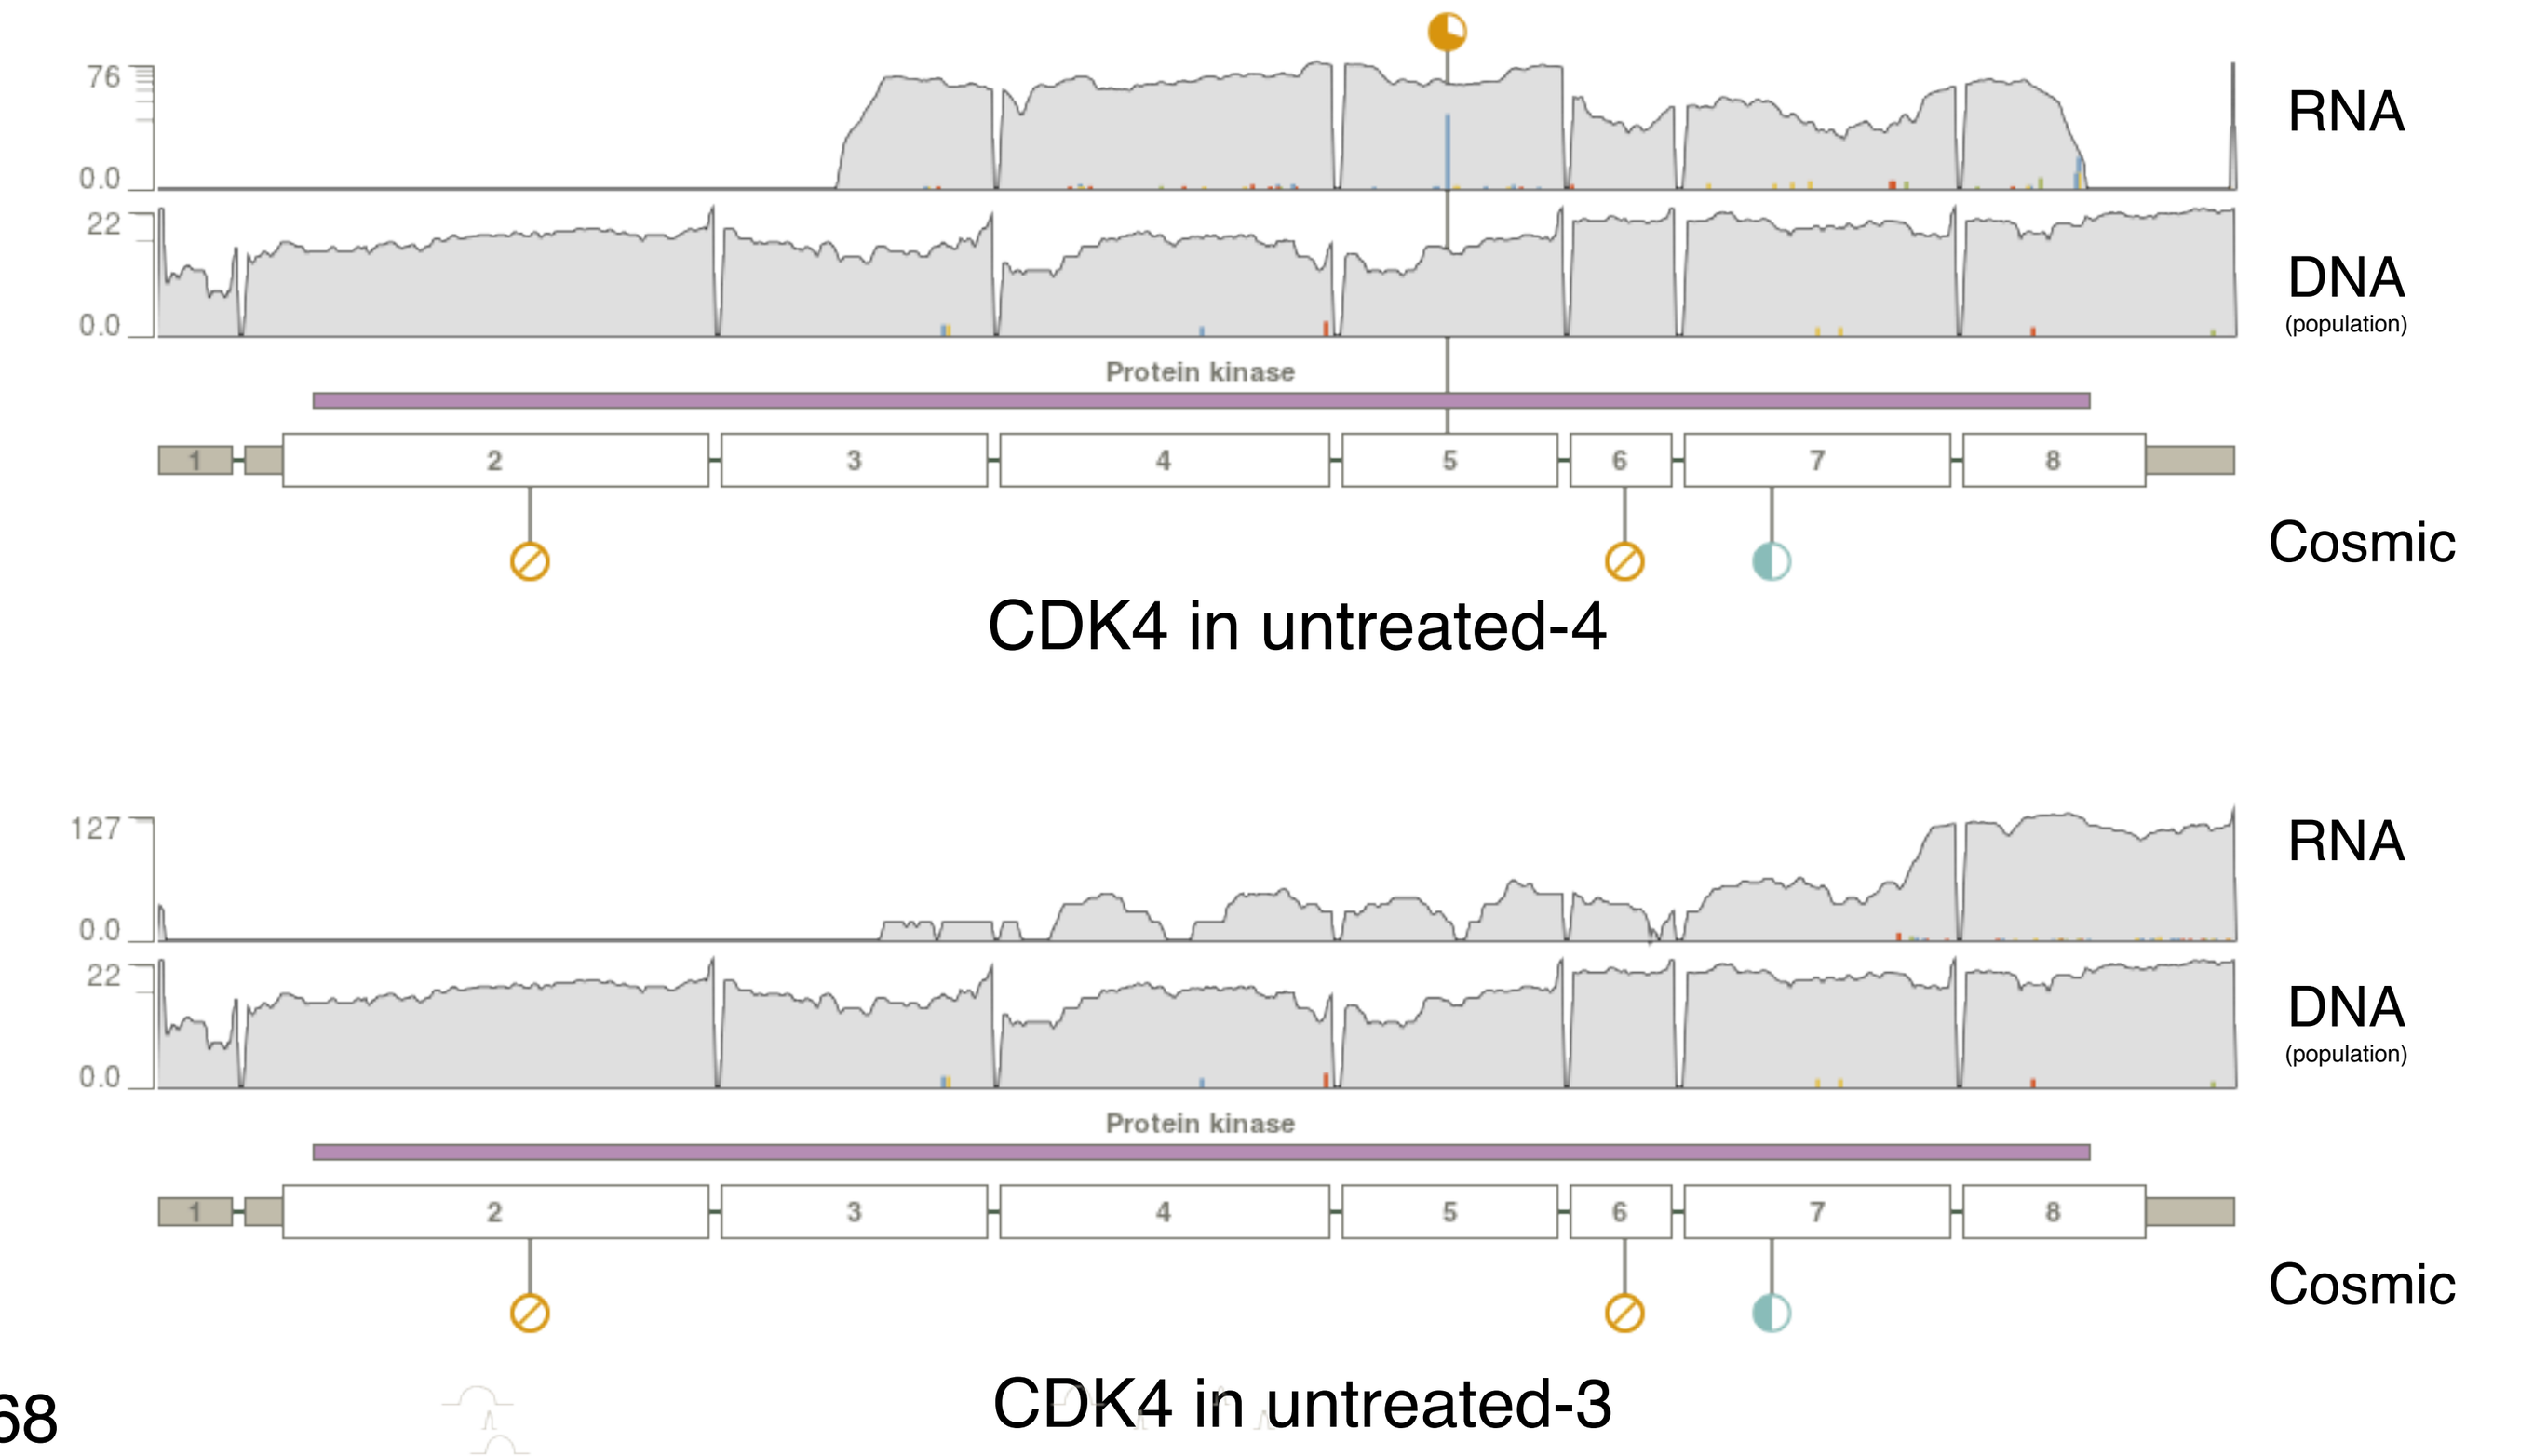
RNA
DNA
(population)
Protein kinase
Cosmic
CDK4 in untreated-4
RNA
DNA
(population)
Protein kinase
Cosmic
CDK4 in untreated-3

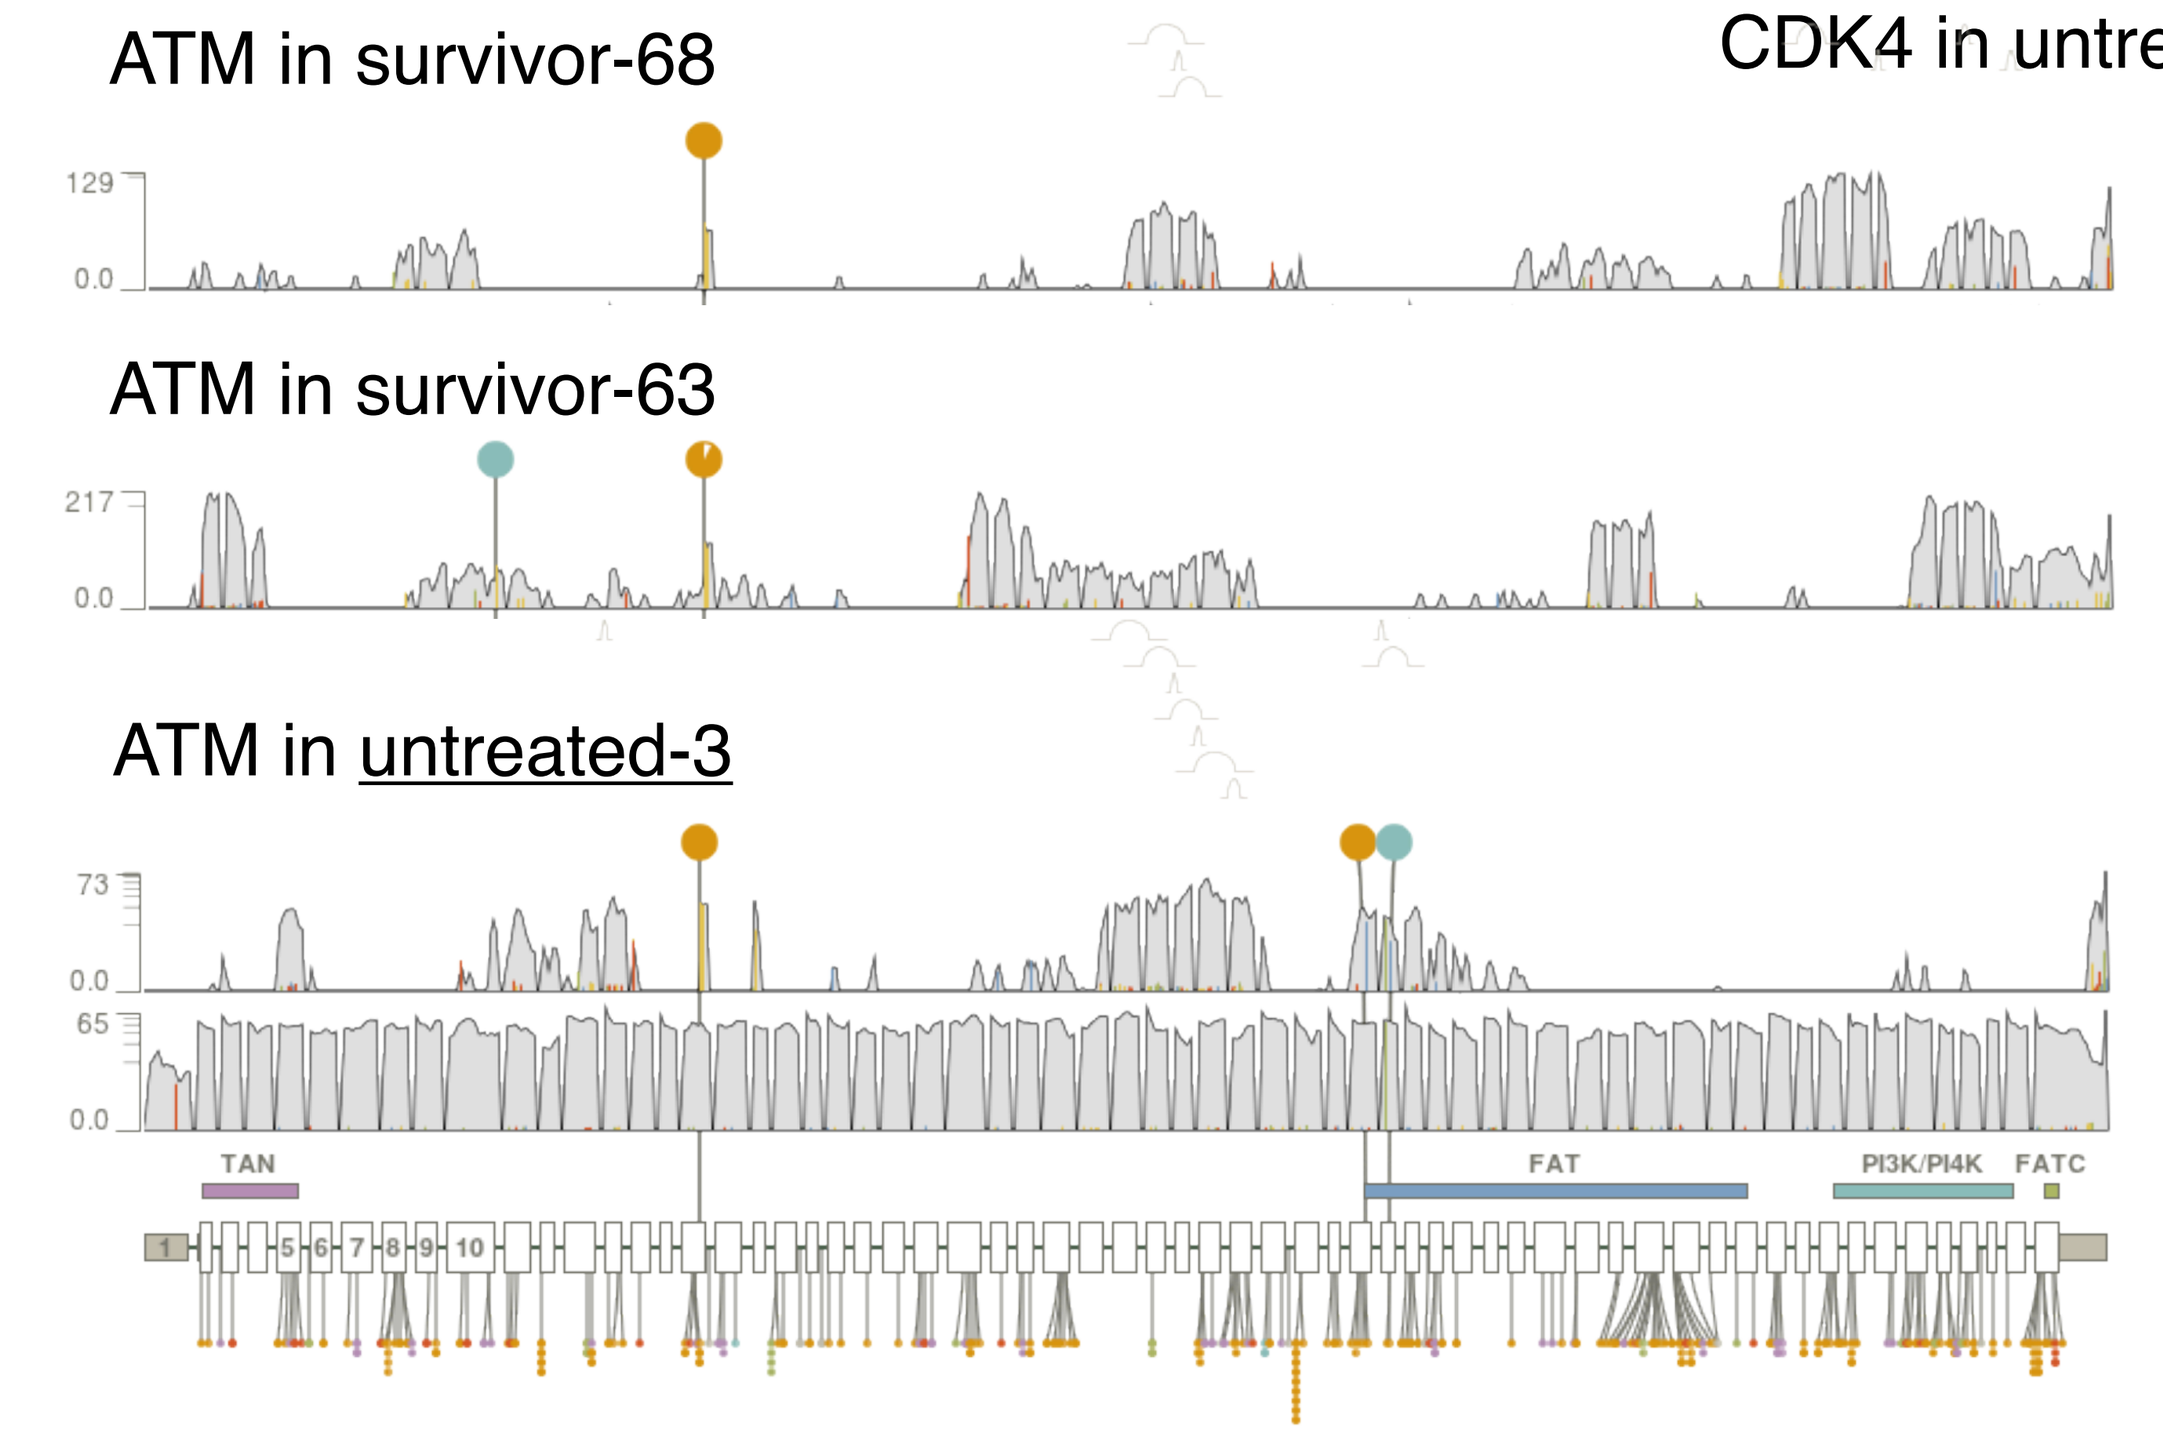
ATM in survivor-68
ATM in survivor-63
ATM in untreated-3
TAN
FAT
PI3K/PI4K
FATC

The coverage of RNA in individual cells, or DNA from a population of cells, is shown for each gene. At each location in the coverage plot, the fraction of non-reference bases is indicated by colored lines. The exon structure and PFAM domains are shown at the bottom, above mutations in the Cosmic database.

Small variant calling

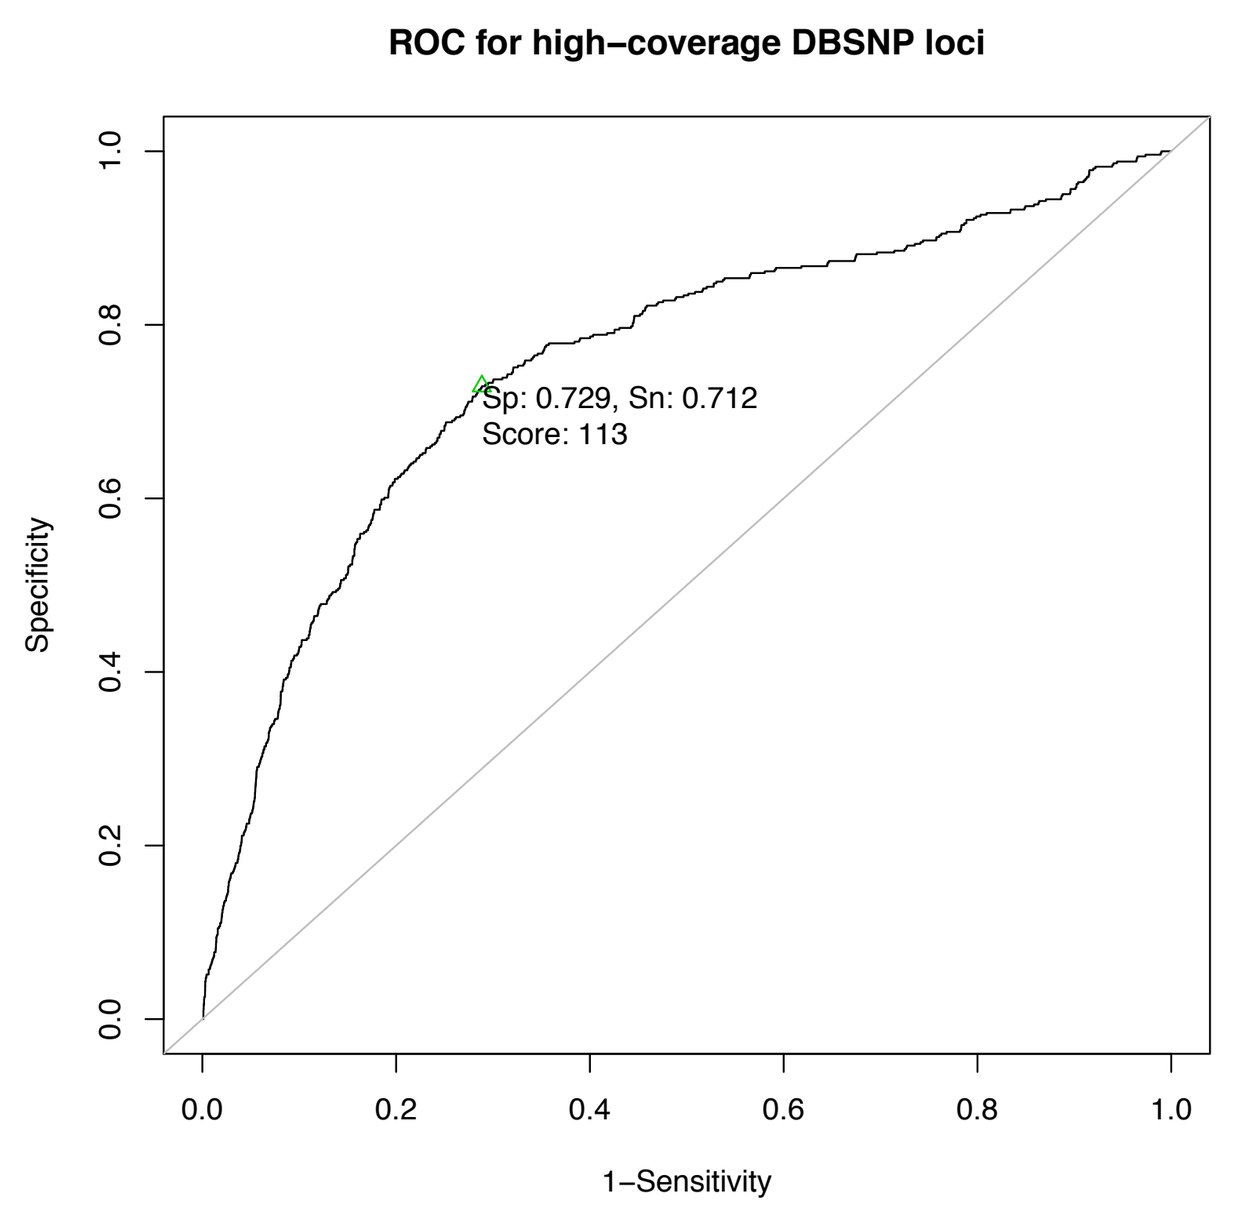
ROC for high–coverage DBSNP loci
Specificity
1–Sensitivity
Sp: 0.729, Sn: 0.712
Score: 113

We find **highly differential mutation rates** within the different treatment conditions, and a dosage effect between low and high doses of paclitaxel. Surviving cells show a variant rate similar to the untreated cells, indicating that survivors either repaired mutations, or experienced fewer mutations initially.

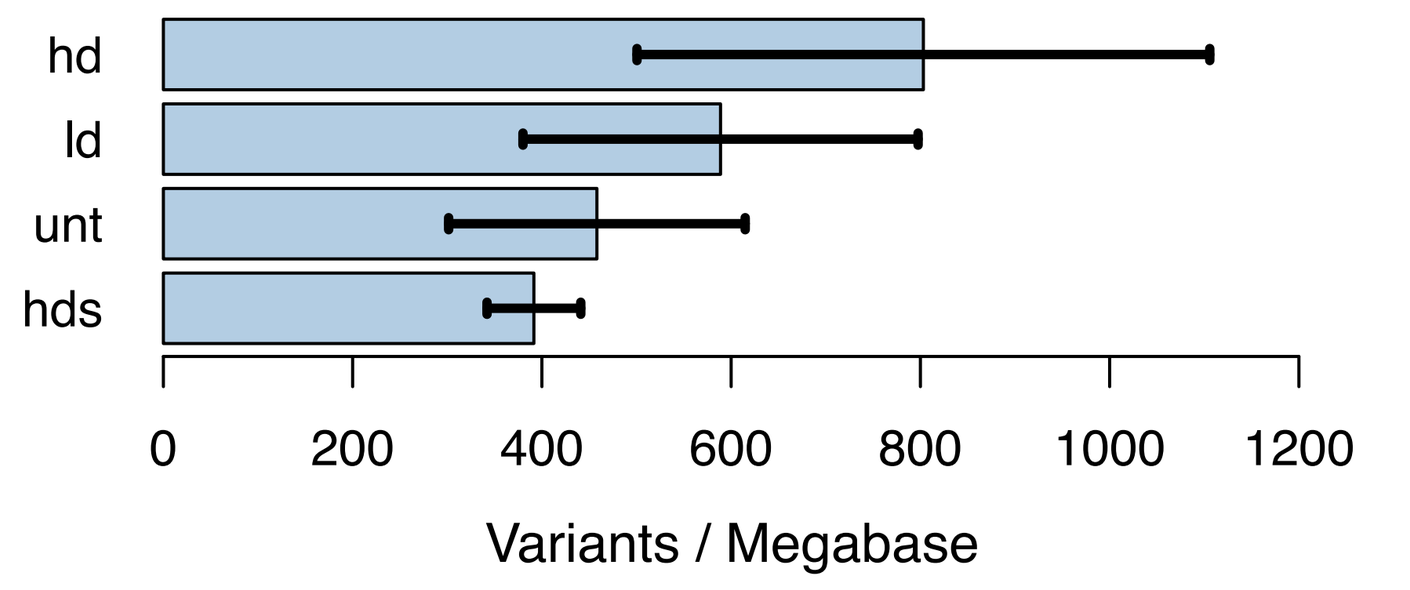
hd
ld
unt
hds
0
200
400
600
800
1000
1200
Variants / Megabase

Using a Bayesian variant caller originally developed for DNA and tumor-normal pairs, we called variants in single-sample mode on both the RNA and on a 10x whole genome DNA sequencing of the tumor population. Using the DNA calls at DBSNP loci as the gold standard, we assessed the sensitivity and specificity of the RNA calls at high-coverage (30x) loci, finding **~70% sensitivity and specificity**, disregarding the confounding effects of RNA editing.

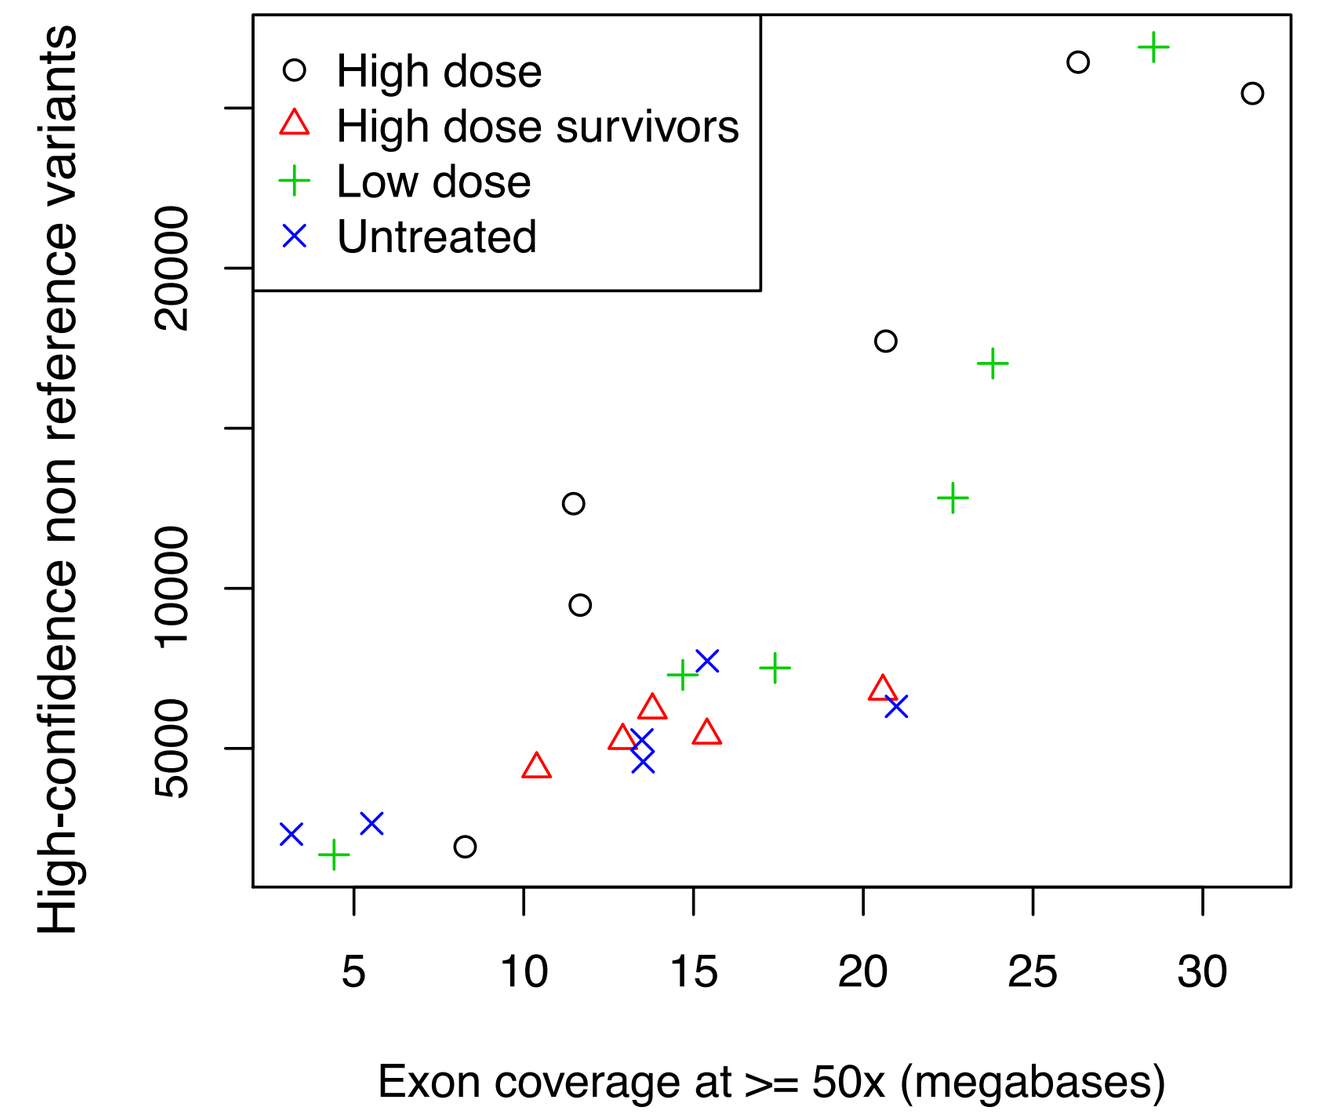
High dose
High dose survivors
Low dose
Untreated
High-confidence non reference variants
Exon coverage at >= 50x (megabases)

Genes mutated in surviving cells

Tubulin
- MAP1A - Microtubule-associated protein 1A
- TTLL5 - Tubulin tyrosine ligase-like family, member 5
- TBCD - Tubulin folding cofactor
- CCT4 - Chaperonin containing TCP1, subunit 4 (delta)

DNA
- CHD1L - Chromodomain helicase DNA binding protein 1-like
- TOX - Thymocyte selection-associated high mobility group box
- NFRKB - Nuclear factor related to kappaB binding protein

Inter-cell
- CPA3 - Carboxypeptidase A3 (mast cell)
- ELFN2 - Extracellular leucine-rich repeat and fibronectin type III
- CTNND1 - Catenin (cadherin-associated protein), delta 1

342 genes were found to have non-silent mutations in at least one of the surviving cells, with no observed variants in the untreated cells' RNA or in population's genomic DNA. Examining genes mutated in at least four of the surviving cells found functional motifs for tubulin related proteins, DNA-binding proteins, and proteins involved in inter-cellular signaling.

This presentation is the intellectual property of the author/presenter. Contact charlie@five3genomics.com for permission to reprint and/or distribute.